Phase I study of the mutant *IDH1* inhibitor ivosidenib: long-term safety and clinical activity in patients with conventional chondrosarcoma

Abstract Number: 11532

William D. Tap[1], Gregory M. Cote[2], Howard Burris[3], Lia Gore[4], Murali Beeram[5], Anthony P. Conley[6], Diego A. Gianolio[7], Zhe Qu[7], Camelia Gliser[7], Susan Pandya[7], Jonathan C. Trent[8]

[1]Memorial Sloan-Kettering Cancer Center, NY, USA, [2]Massachusetts General Hospital Cancer Center, MA USA, [3]Sarah Cannon Research Institute, TN, USA,[4]University of Colorado Cancer Center, Aurora, CO, USA,[5]START Center for Cancer Care, San Antonio, TX, USA, [6]MD Anderson Cancer Center, TX, [7]Servier Pharmaceuticals, MA USA , [8]Sylvester Comprehensive Cancer Center, FL, USA

INTRODUCTION

- Conventional chondrosarcomas (CS) make up 90% of CS and are rare primary bone malignancies, for which there are no approved systemic therapies.[1]
- Surgical resection is the primary treatment and can result in morbidity (e.g., limb amputations, limitations in mobility), and approximately 25% of patients will experience local recurrence after surgery.[2–3]
- In the metastatic disease setting, the 5 years overall survival (OS) is 5%.[4–6]
- Mutations in the isocitrate dehydrogenase 1 (*IDH1*) gene occur in approximately 50% of conventional CS.[7]
- Ivosidenib (IVO) is a potent oral targeted inhibitor of mutant *IDH1*, approved as a 500 mg daily dose in the US and EU in *IDH1* mutated (m*IDH1*) AML and m*IDH1* cholangiocarcinoma.
- In a phase 1 study (NCT02073994), at the data cut-off of 16 January 2019, in 21 patients with m*IDH1* advanced CS including 13 with the conventional sub-type, IVO demonstrated manageable toxicity (without dose-limiting toxicities), suppression of the oncometabolite 2-hydroxyglutarate (2-HG) by 56.7% (range 14%–94.2%) at IVO dose levels ≥300 mg/day, and disease control rate of 52.4%, with a median progression-free survival (PFS) of 5.6 months.[8]
- Here we report long term safety and efficacy results of IVO among the conventional CS population based on the data cut-off date of 15 September 2022.

OBJECTIVE

- To assess safety, tolerability and efficacy of IVO in patients with m*IDH1* conventional chondrosarcoma patients using long-term follow up data.

METHODS

- The methodology of this phase I multicenter open-label dose-escalation and expansion study has previously been published in detail and will be summarized here briefly.[8]
- For dose escalation, IVO was administered orally in continuous 28-day cycles at 100 mg twice daily (BID) and 300, 400, 500, 600, 800, 900, and 1,200 mg QD using a standard 3+3 design.
- Included patients had known mutant *IDH1* advanced CS, and with other known co-mutations, that had progressed or not responded to prior therapy.
- Primary outcome was safety and tolerability
- Secondary outcomes included clinical activity (objective response rates [ORR, defined as rate of complete response (CR) + partial response (PR)], stable disease [SD] and PFS).
- Adverse events (AEs) were assessed and reported per the Common Terminology Criteria for Adverse Events (CTCAE version 4.03) and responses were assessed every other cycle using Response Evaluation Criteria in Solid Tumors (RECIST version 1.1).

RESULTS

- This trial was initiated in March 2014. 168 patients with m*IDH1* advanced solid tumors received IVO. Of these, 21 had CS; 13 had conventional CS, 6 had dedifferentiated histology, and 2 were unknown. Data cut-off of this analysis is 15 September 2022.
- Baseline characteristics of the 13 patients with conventional CS are summarised in **Table 1**.

Table 1: Baseline characteristics of patients with conventional CS

Characteristics	N (%) unless otherwise stated
Age, years, median (range)	54.0 (38, 88)
Gender	
Male	9 (69.2)
Female	4 (30.8)
AJCC Tumor Grade (at screening)	
I	2 (15.4)
II	8 (61.5)
III	1 (7.7)
Unknown	2 (15.4)
ECOG performance score at baseline	
0	7 (53.8)
1	6 (46.2)
Previous lines of systemic therapy	
0	7 (53.8)
1	4 (30.8)
≥2	2 (15.4)
Previous radiotherapy	3 (23.1)

- Doses received were 100 mg IVO BID [n=1], 400 mg QD [n=1], 500 mg QD [n=7], 800 mg QD [n=2], and 1200 mg QD [n=2].
- 2-HG was maximally suppressed at the 500 mg dose. Details on dosing, pharmacokinetics and pharmacodynamics of IVO have been previously published.[8,9]
- Median number of doses was 341 (range: 14-2801); median relative dose intensity was 100%.
- Median treatment duration was 11.3 months (range: 0.5-92.6 months; **Figure 1**) and mean (SD) treatment duration was 31.9 months (36.2).
- Six patients (46.2%) continued on IVO for at least one year: 3/7 in the 500 mg QD group, 1/2 in the 800 mg QD group and 2/2 in the 1200 mg QD group; four patients (30.8%) continued therapy for >6 years (two of which were treated for >7 years).
- Of the 8 patients with baseline co-mutation data, 5 had no co-mutation and 3 had detected co-mutations (range 1-3).

Figure 1: Treatment duration of patients with conventional CS

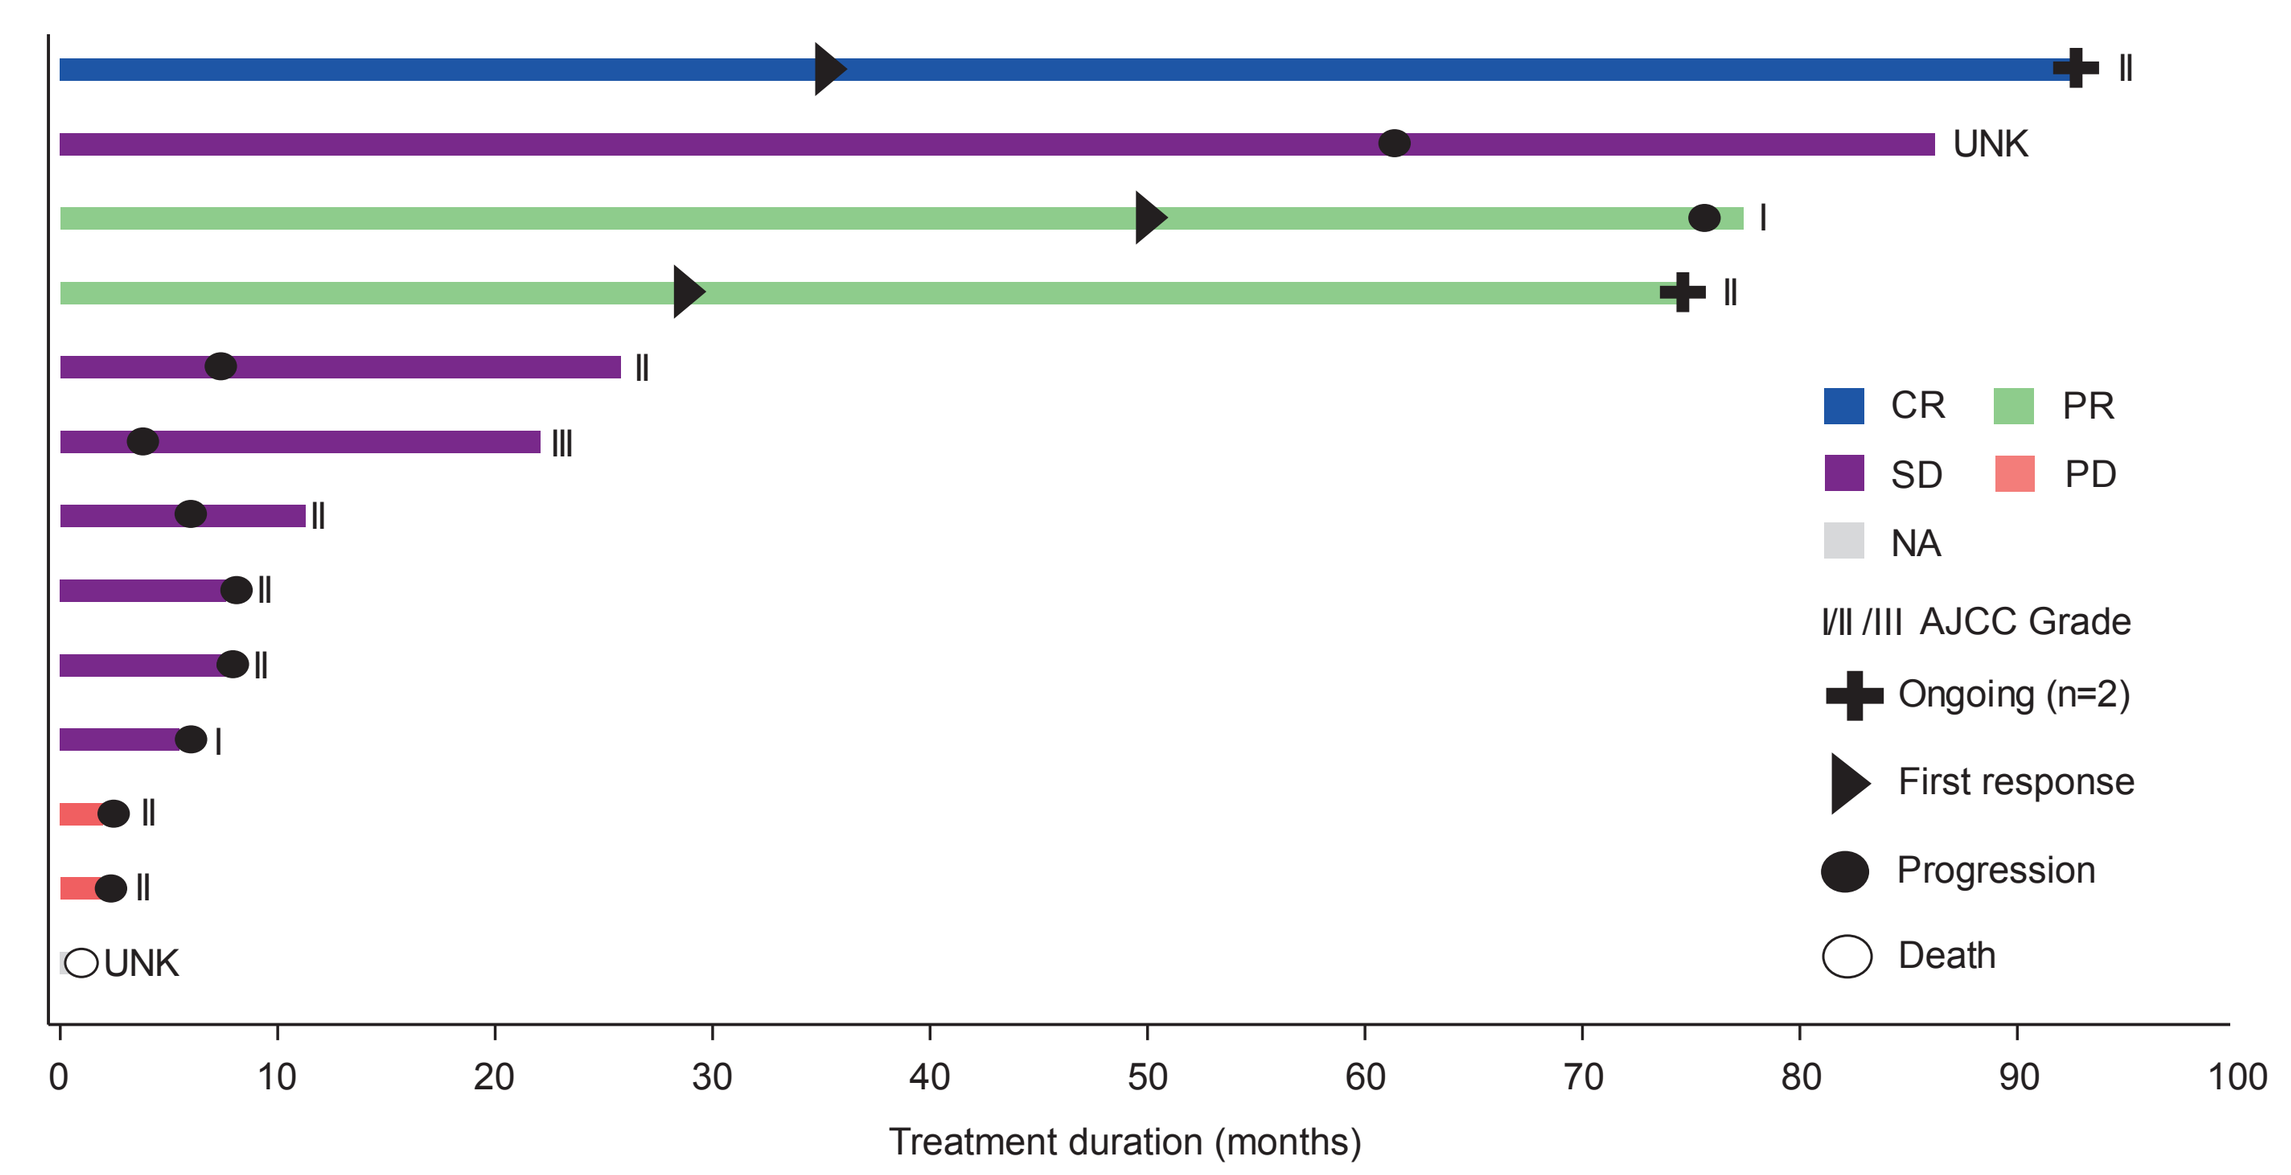
II
UNK
I
II
II
III
II
II
II
I
II
II
UNK
CR
PR
SD
PD
NA
I/II /III AJCC Grade
Ongoing (n=2)
First response
Progression
Death
0
10
20
30
40
50
60
70
80
90
100
Treatment duration (months)

CR= complete response, PD= progressive disease, PR= partial response, SD= stable disease, NA= not assessed. UNK=unknown. As per RECIST v1.1, SD occurring with <42 days of the first dose is assigned as UNK. AJCC Grade at screening

ACKNOWLEDGEMENTS & DISCLOSURES

We acknowledge and thank all patients and their families who took part in the studies, all investigators and study team, and Ty Subhawong (University of Miami Miller School of Medicine, Miami, USA) for interpreting and supplying the radiographic images. This study was supported by Agios Pharmaceuticals, Inc. Servier Pharmaceuticals LLC has completed the acquisition of Agios' oncology business. **DG**, **ZQ**, **CG** and **SP** are employees of Servier Pharmaceuticals. **LG** holds the Ergen Family Chair in Pediatric Oncology at Children's Hospital Colorado and is supported by P30 CA46934. Editorial assistance was provided by Emily Eagles of Empowering Strategic Performance Ltd, supported by Servier Pharmaceuticals LLC.

RESULTS

Safety and tolerability

- In total, 13 patients experienced AE's, with 9 having AEs that were treatment related. One grade ≥3 AE (hypophosphatemia) was possibly related to treatment; this AE resolved and did not require IVO dose adjustment.
- The most frequent all grade treatment emergent AEs (mostly grade 1 or 2) were diarrhea (n=5) and nausea (n=5; **Table 2**).
- Six patients had grade ≥3 AEs and three patients experienced four serious AEs (infective cholangitis, pneumonia, wound infection, fall) however no serious AE was considered related to treatment.
- There were no discontinuations, dose reductions or deaths due to AEs.

Table 2: Most frequent* AEs (all grades) in patients with conventional CS

Adverse event (in ≥20% of patients)	N (%) patients
Diarrhea	5 (38.5)
Nausea	5 (38.5)
Arthralgia	4 (30.8)
Constipation	4 (30.8)
Electrocardiogram QT prolonged	4 (30.8)
Fatigue	4 (30.8)
Pain in extremity	4 (30.8)
Upper respiratory tract infection	4 (30.8)

* Occurring in ≥3 patients

Efficacy

- The ORR was 23.1%.
- Median duration of response was 42.5 months (range: 25.8-51.8 months).
- One patient with a base of the skull lesion achieved a complete response (1200 mg IVO QD); two achieved partial response (one 500 mg and one 1200 mg IVO QD); seven had stable disease (five received ≥500 mg IVO QD) and two had progressive disease (both received ≥500 mg IVO QD; **Figure 2**).
- All responses occurred after >2 years on treatment.
- Median PFS was 7.4 months (95% CI: 2.0-61.3) (**Figure 3**).
- The PFS rate at 6 months was 53.8% and at 12 months was 30.8%.

Figure 2: Best percentage change from baseline in target lesion measurement in patients with conventional CS

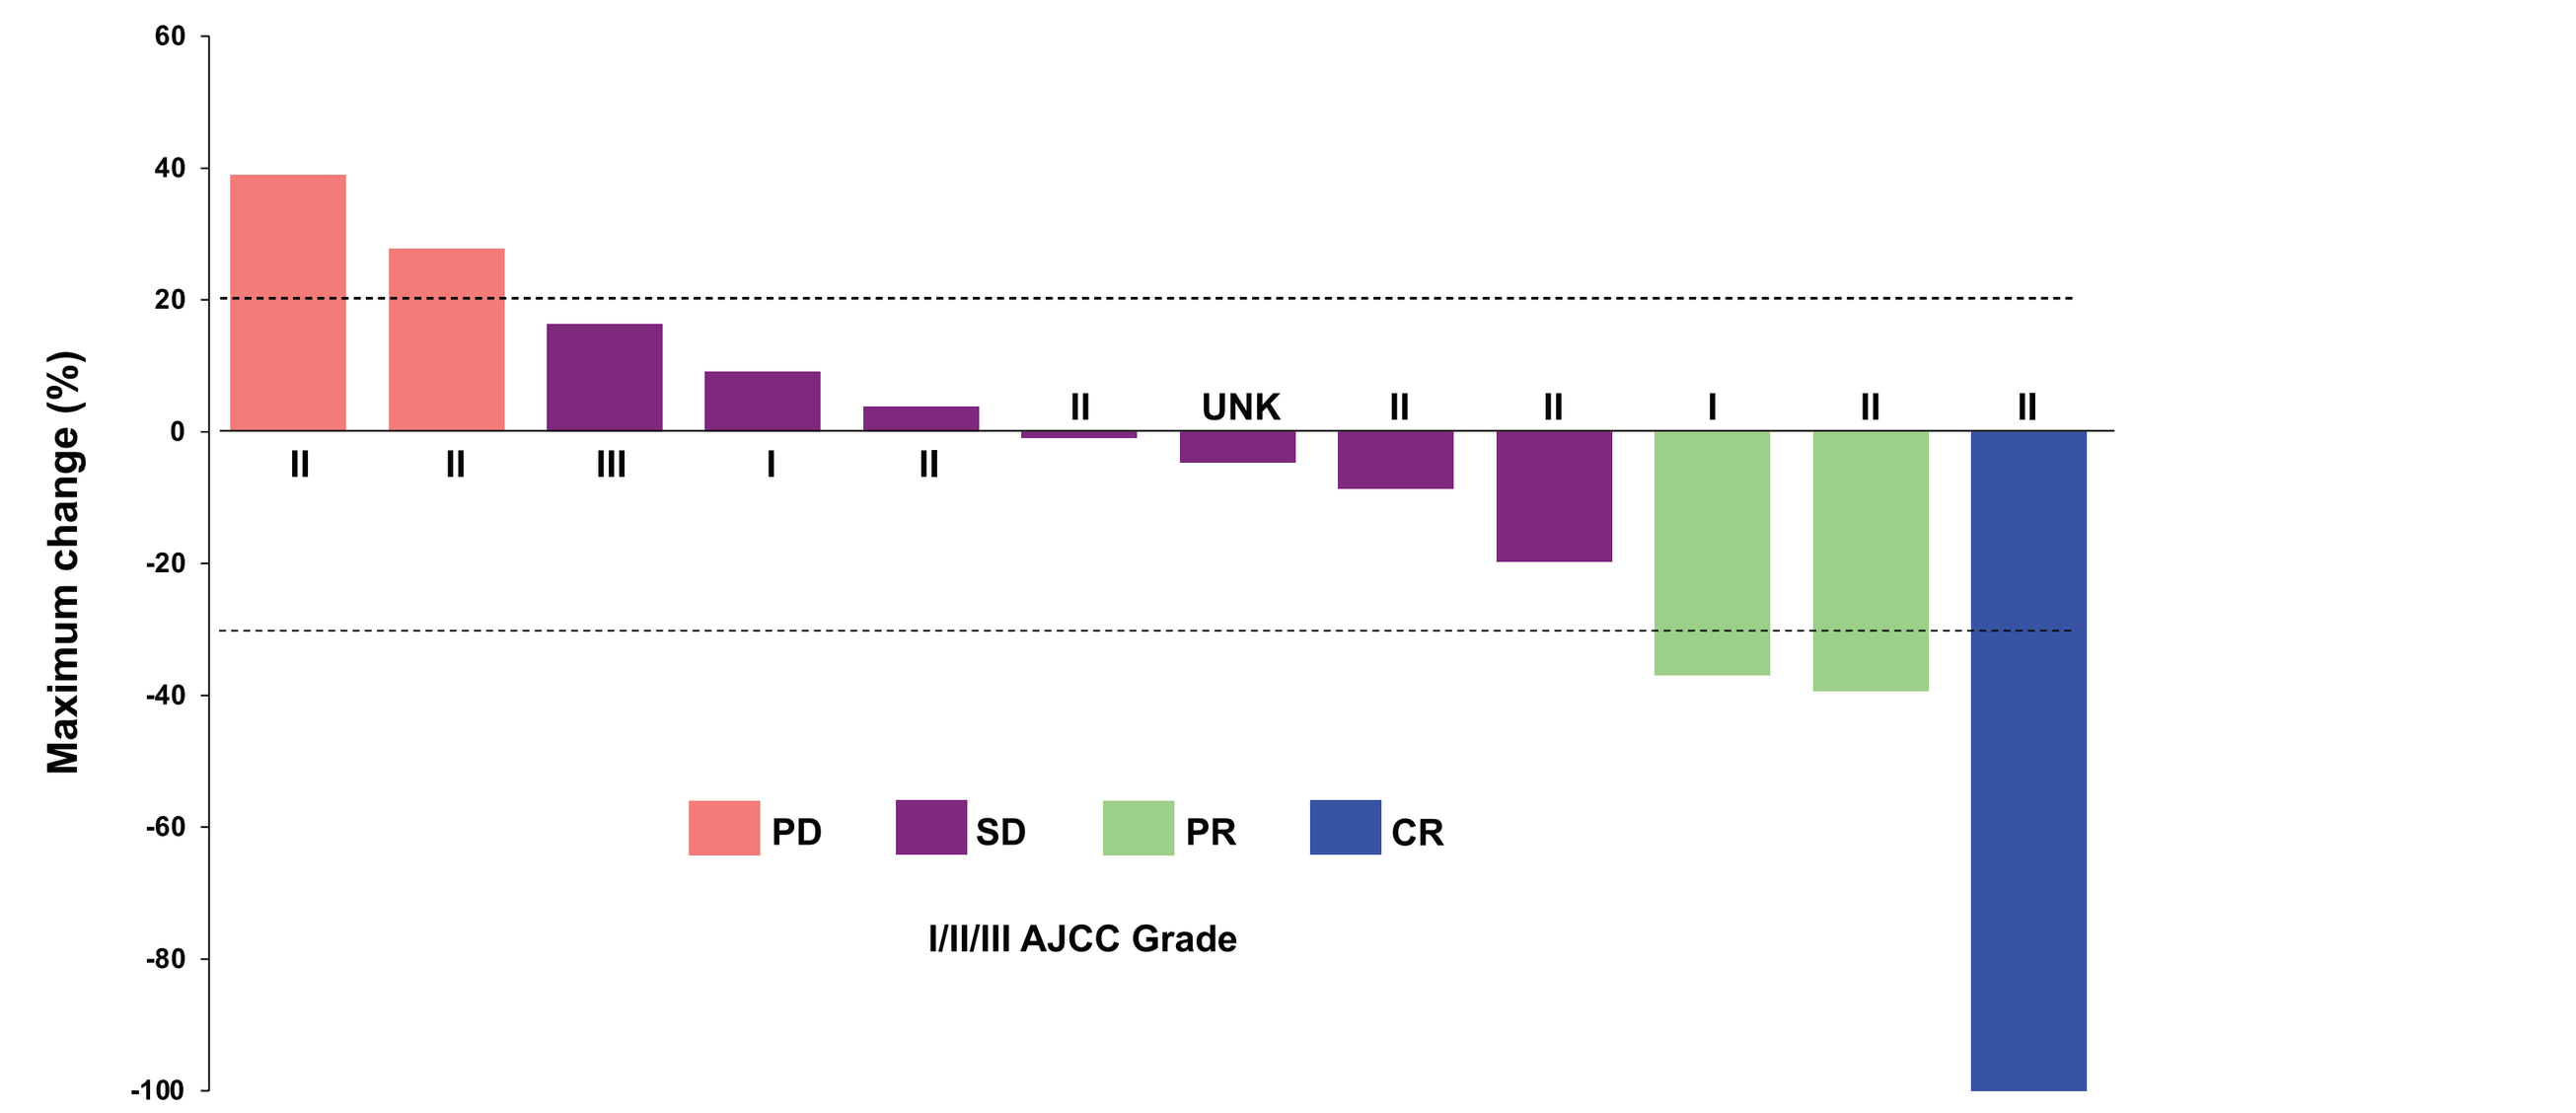
60
40
20
0
-20
-40
-60
-80
-100
Maximum change (%)
II
II
III
I
II
II
UNK
II
II
I
II
II
PD
SD
PR
CR
I/II/III AJCC Grade

CR= complete response, PD= progressive disease, PR= partial response, SD= stable disease. As per RECIST v1.1, SD occurring with <42 days of the first dose is assigned as UNK. One patient was not evaluable. UNK=unknown. As per RECIST v1.1, SD occurring with <42 days of the first dose is assigned as UNK. AJCC Grade at screening.

REFERENCES

1. NCCN Guidelines Version 2.2023 Chondrosarcoma
2. Aggerholm-Pedersen et al. Orth Res Rev. 2019, 11:109-116
3. Andreou et al. Acta Orthop. 2011, 82:749-755
4. Fromm et al. BMC Canc. 2018, 18:849
5. Chen et al. J Chin Med Assoc. 2020, 83:669-673
6. Fiorenza et al, J Bone Joint Surg Br. 2002, 84:93-99
7. Amary et al. J Pathol 2011, 224;334
8. Tap et al. J Clin Oncol. 2020, 38(15):1693-1701
9. Fan, et al. Invest New Drugs 2020, 38:433-444

RESULTS

Figure 3: Kaplan-Meier curve for PFS in patients with conventional CS

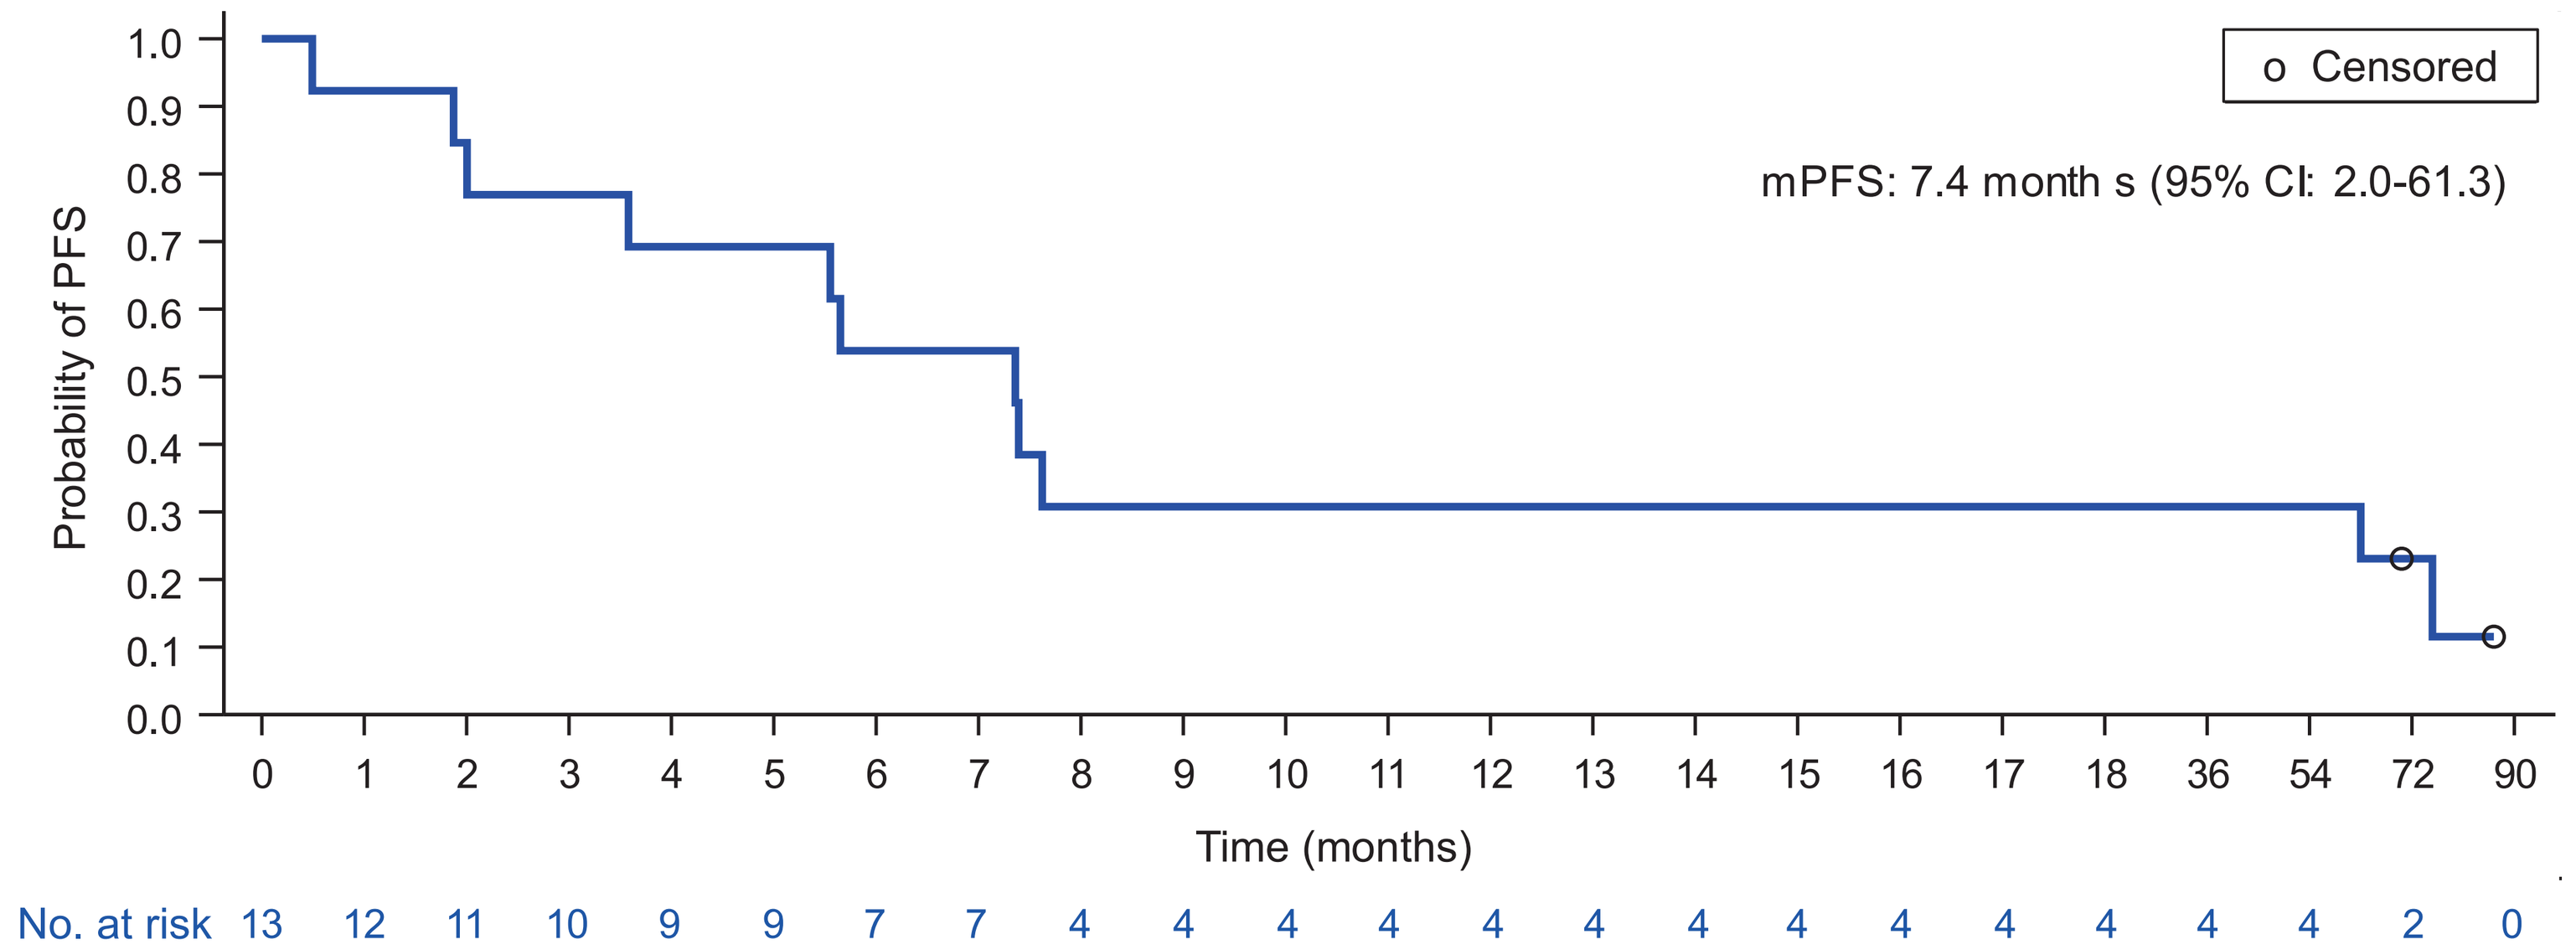
o Censored
mPFS: 7.4 month s (95% CI: 2.0-61.3)
Probability of PFS
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 36 54 72 90
Time (months)
No. at risk 13 12 11 10 9 9 7 7 4 4 4 4 4 4 4 4 4 4 4 4 4 2 0

- Imaging of a 38-year-old female with Grade 1 conventional CS (involving the right pelvis) previously treated with one systemic therapy who then received IVO is shown in **Figure 4.**
- Coronal fat suppressed T2-weighted MRI of the pelvis demonstrated the lobular CS in the right iliac and ischium, with relatively uniform hyperintensity and a lateral extraosseous component (yellow arrow) at baseline (**Figure 4**).
- After treatment, there was reconstitution of fatty marrow signal in the right iliac, with smaller residual islands of T2 hyperintense foci (green arrows) and regression of the extraosseous component, resulting in PR (**Figure 4**) that lasted 25.8 months.

Figure 4: Radiographic changes in a patient with conventional CS with a PR

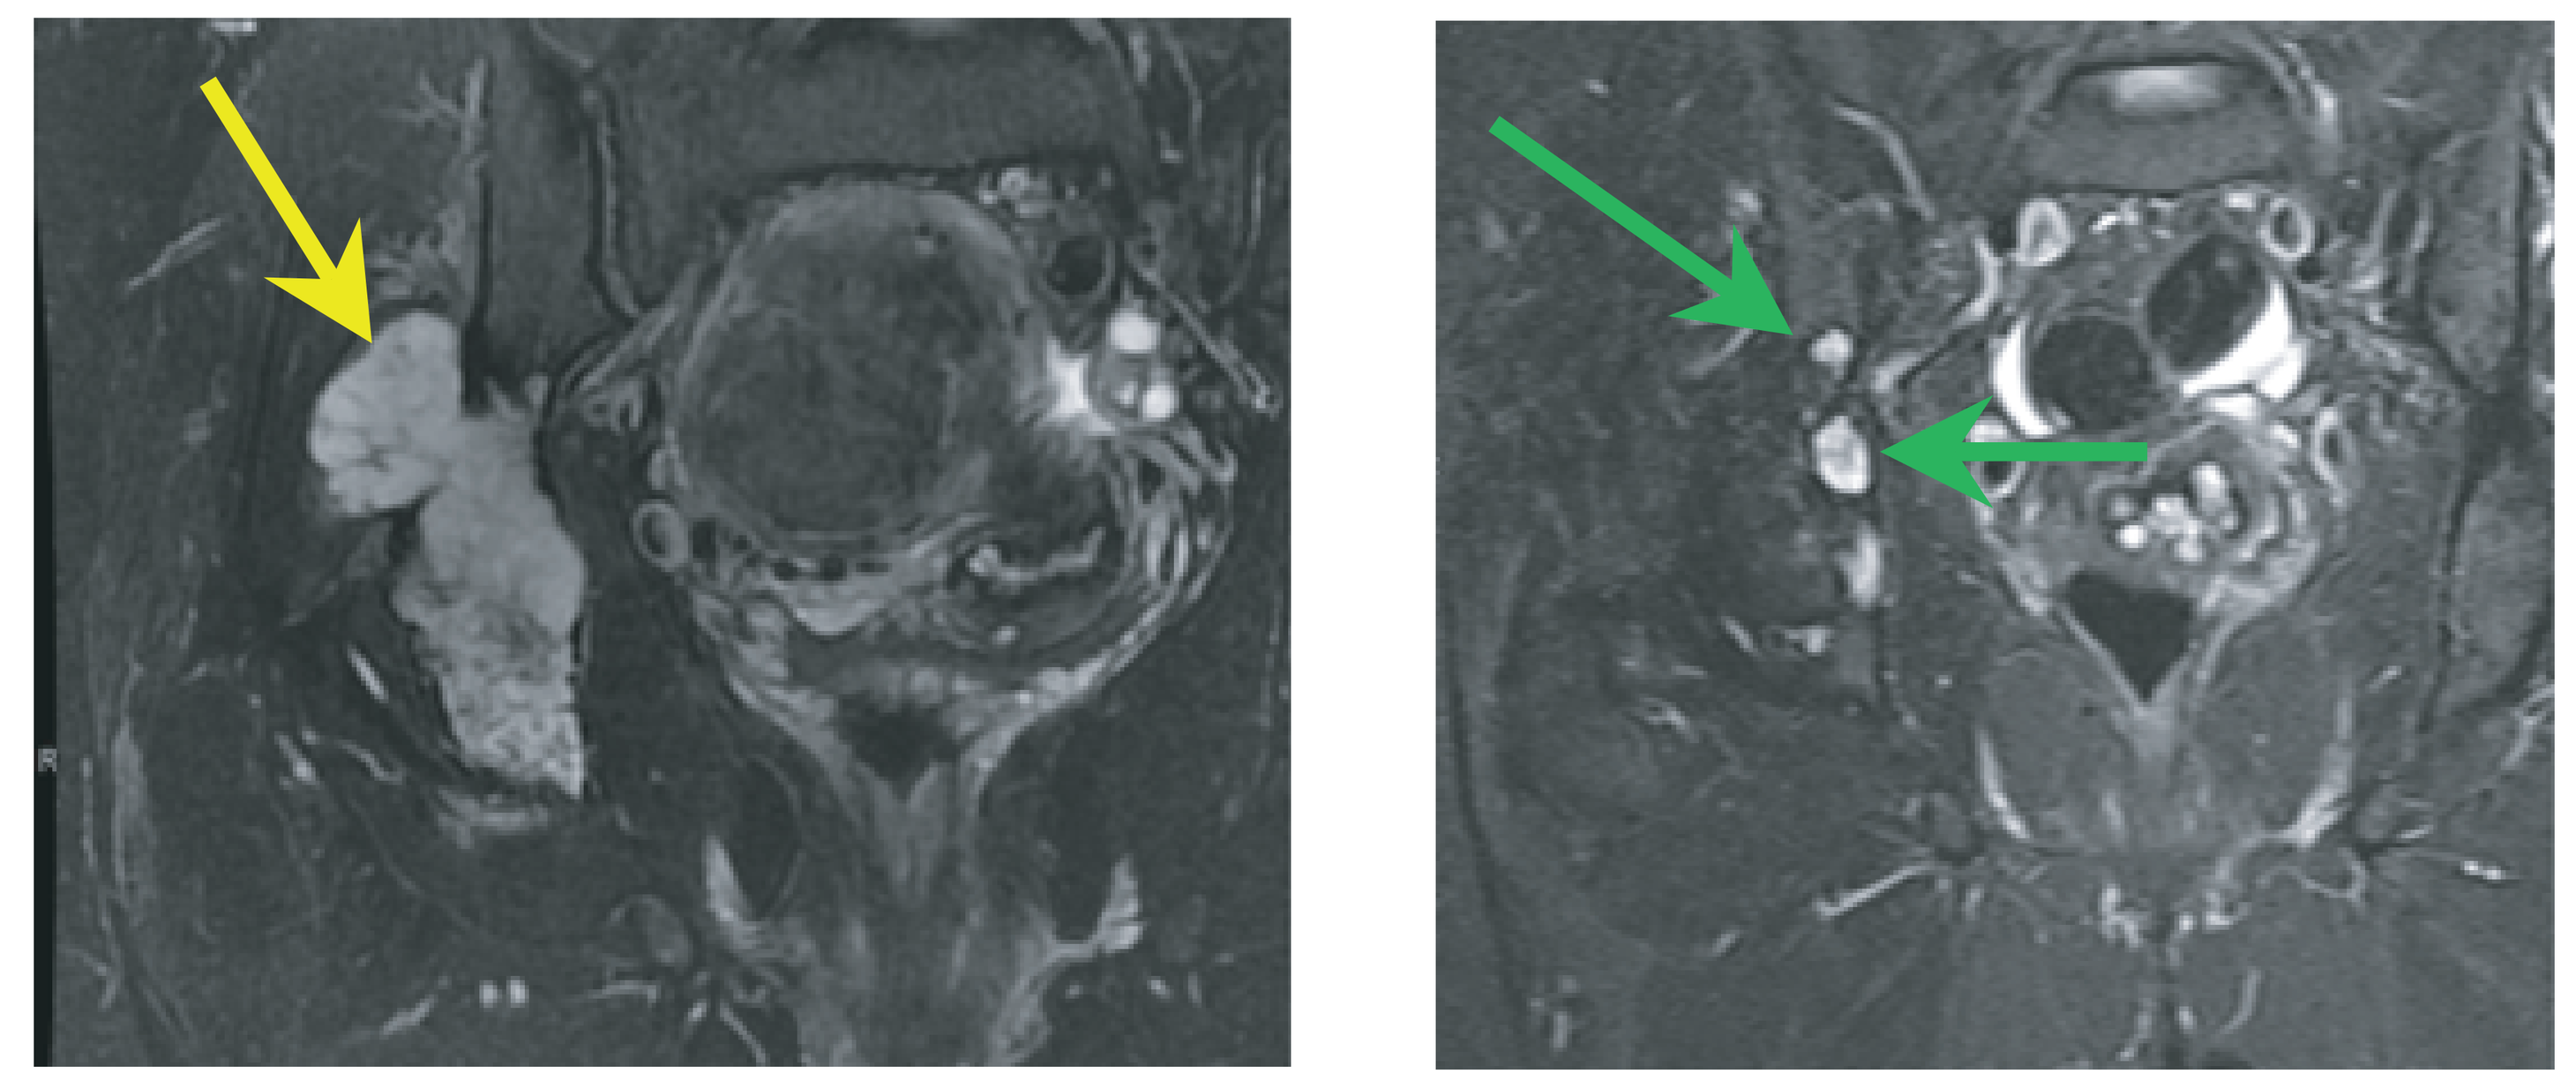

Baseline **Cycle 55**

CONCLUSIONS

- In patients with m*IDH1* advanced conventional CS (n=13), IVO demonstrated manageable toxicity with mostly grade 1 or 2 treatment emergent AEs.
- Disease control was durable for patients with advanced conventional CS including long-term responses. Treatment duration was >6 years for 4 patients (30.8%), and >7 years for 2 (15.4%) patients.
- Median PFS for advanced conventional CS patients was 7.4 months. ORR was 23.1% including 2 partial responses and 1 complete response.
- Genomic analysis was incomplete but revealed known co-mutations.
- Future studies with broader exploratory genomic sequencing efforts are warranted to confirm the promising efficacy of IVO in patients with advanced m*IDH1* conventional CS.

Presented at the American Society of Clinical Oncology (ASCO), June 2–6, 2023, Chicago, IL